Stability of Reconstituted Venom Extracts at Room and Elevated Temperatures

Liza Garriques and Shannon Brown, Jubilant HollisterStier (Spokane, WA)

AAAAI
2026
ID: 853

Background

Hymenoptera Venom Extracts, which are indicated for allergy diagnosis and immunotherapy, are provided as freeze-dried products, then reconstituted to a target strength prior to use.

The stability profiles for storage at recommended conditions (2-8°C) are well-established and support the expiration dates stated in the product package insert for 100 µg/mL reconstituted venom.[1] However, the stability of the 100 µg/mL reconstituted venom, when exposed to temperatures above 2-8°C, is not routinely monitored.

Phospholipase A (PLA) and Hyaluronidase (HLA) are two important allergenic components of Hymenoptera venom products. Measurements of PLA and HLA enzyme activity are required by FDA for lot release and stability testing.

Purpose

This study sought to characterize the stability of 100 µg/mL reconstituted venoms at room (RT) and elevated (ET) temperatures.

References:

1. HollisterStier Allergy Package Insert 355125-H04, *Instructions and Dosage Schedule for Allergenic Extracts – Hymenoptera Venom Products*

Methods

<u>HollisterStier Standardized Venom Products</u>

- Honey Bee, Item No. 6781PG (HB)
- Wasp, Item No. 6784PG3 (W)
- White Faced Hornet, Item No. 6782PG (WFH)
- Yellow Jacket, Item No. 6785PG (YJ)
- Yellow Hornet, Item No. 6783PG (YH)

<u>Diluent</u>: Sterile Albumin Saline with Phenol (ABS)

<u>Tests Methods</u>

- Phospholipase (PLA) Activity by Radial Diffusion Assay
- Hyaluronidase (HLA) Activity by Radial Diffusion Assay
- Bioburden (BB) – detection of viable aerobic organisms by Membrane Filtration method

<u>Procedure</u>

- Freeze-dried venom products reconstituted to 100 µg/mL with ABS, per Package Insert.
- Reconstituted venom vials of all types stored in a controlled room temperature (25°C) chamber for up to 30 days. Vials of YH also stored in a controlled elevated temperature (40°C) chamber for up to 30 days.
- Samples pulled at time-points throughout study and tested for PLA and HLA activity. Bioburden tested at last time-point.
- Trended test results with time and compare to stability specification.

Results

Fig. 1 – PLA Activity for HB and Wasp

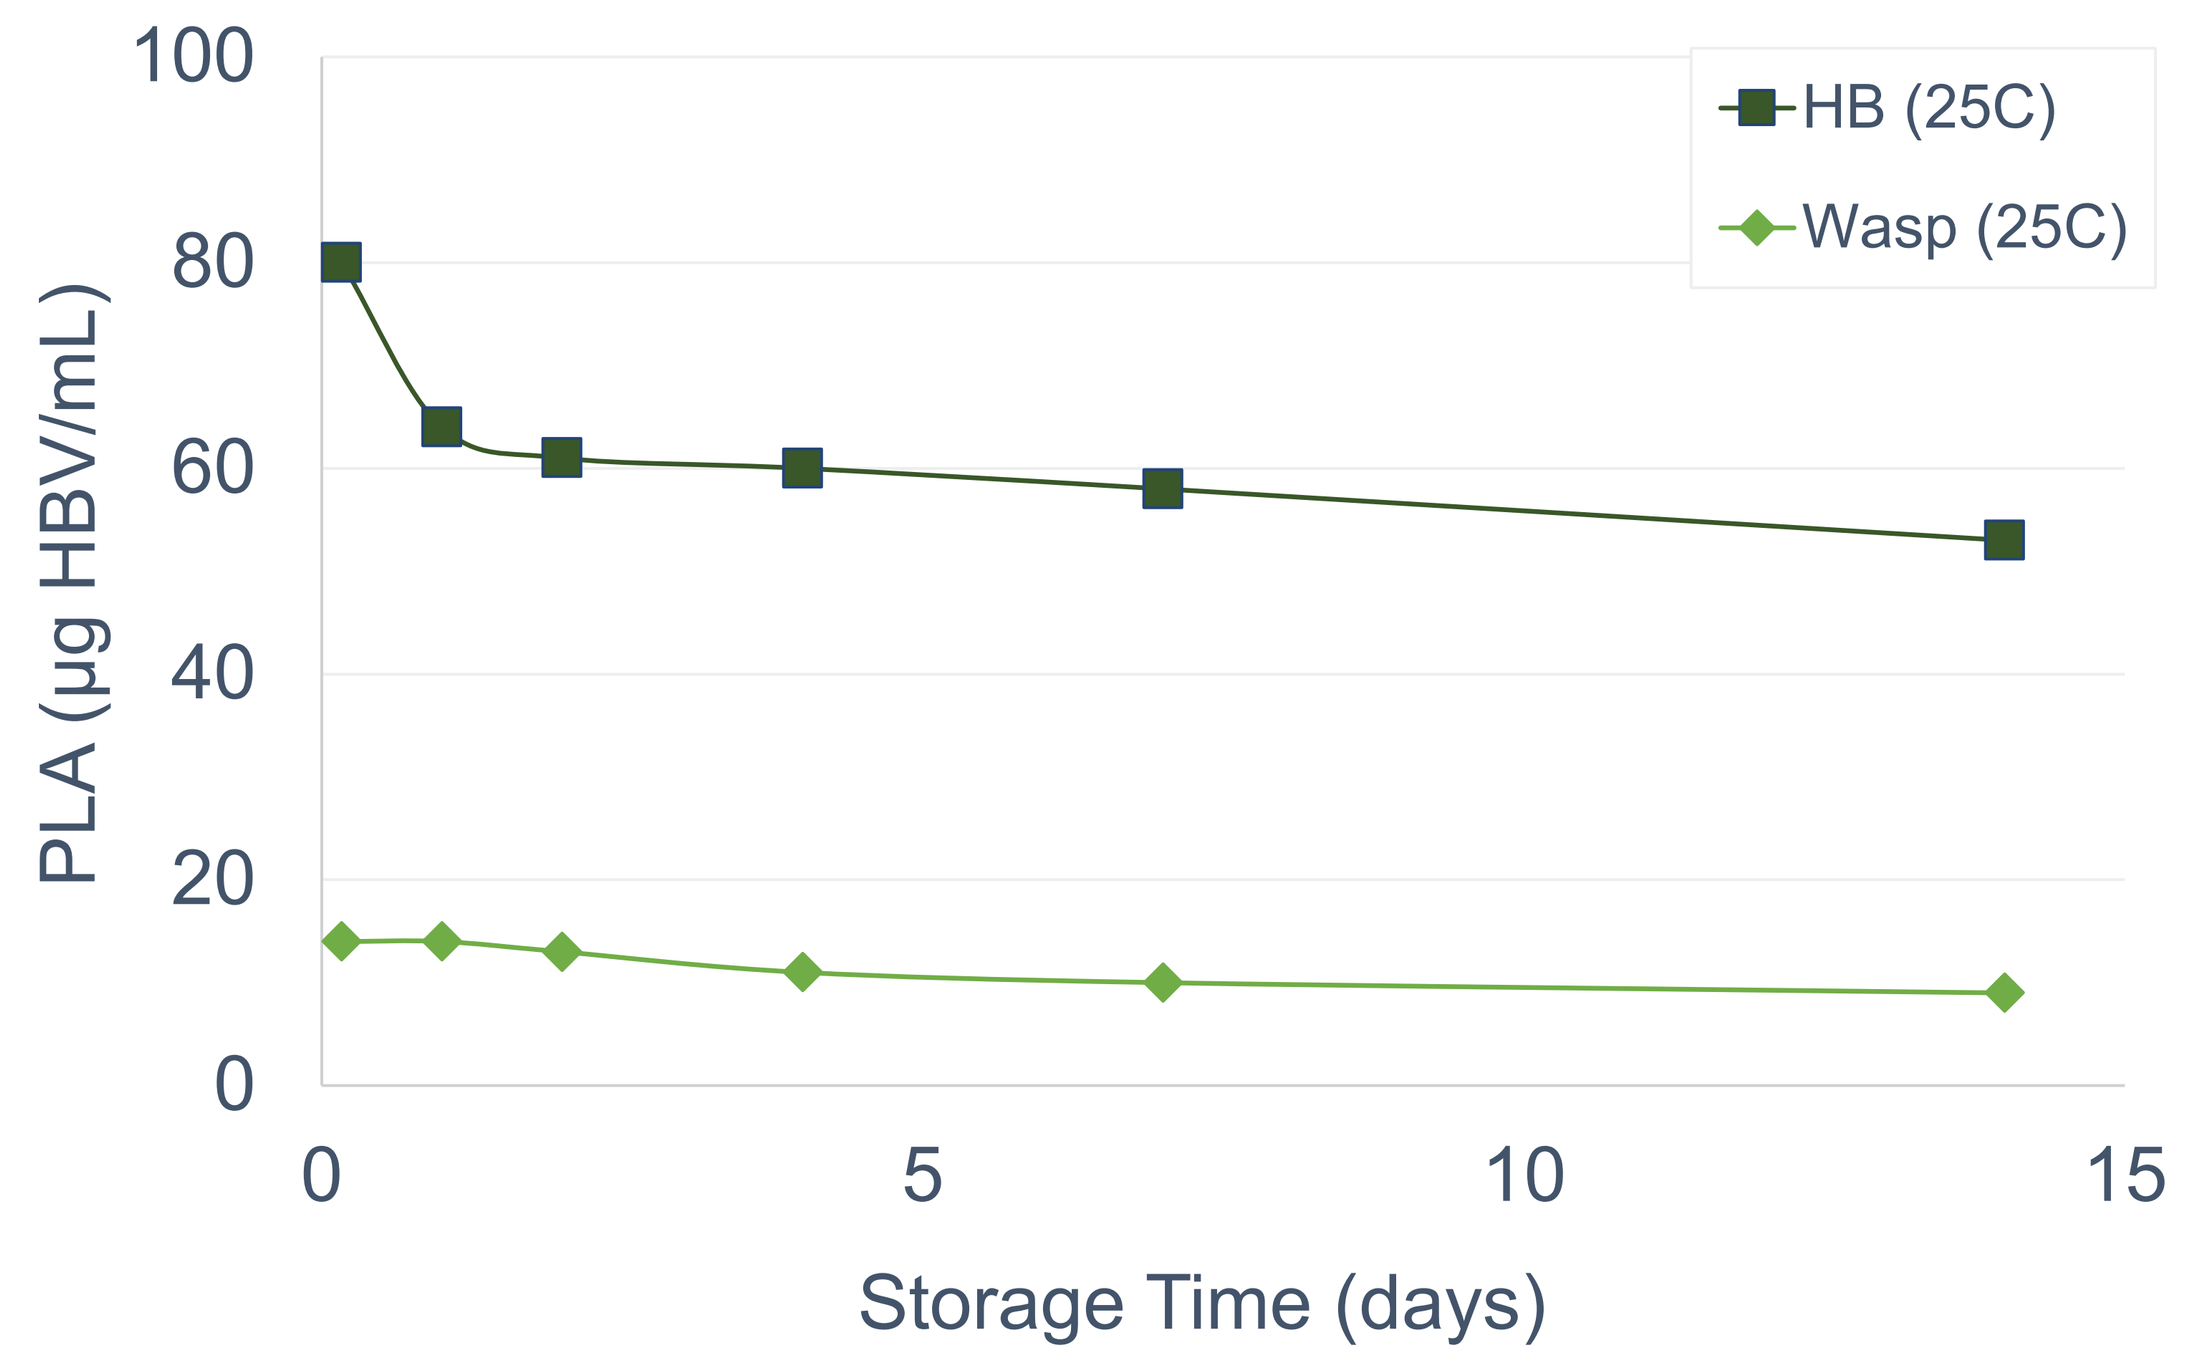
PLA (µg HBV/mL)
Storage Time (days)
HB (25C)
Wasp (25C)

Fig. 2 – HLA Activity for HB and Wasp

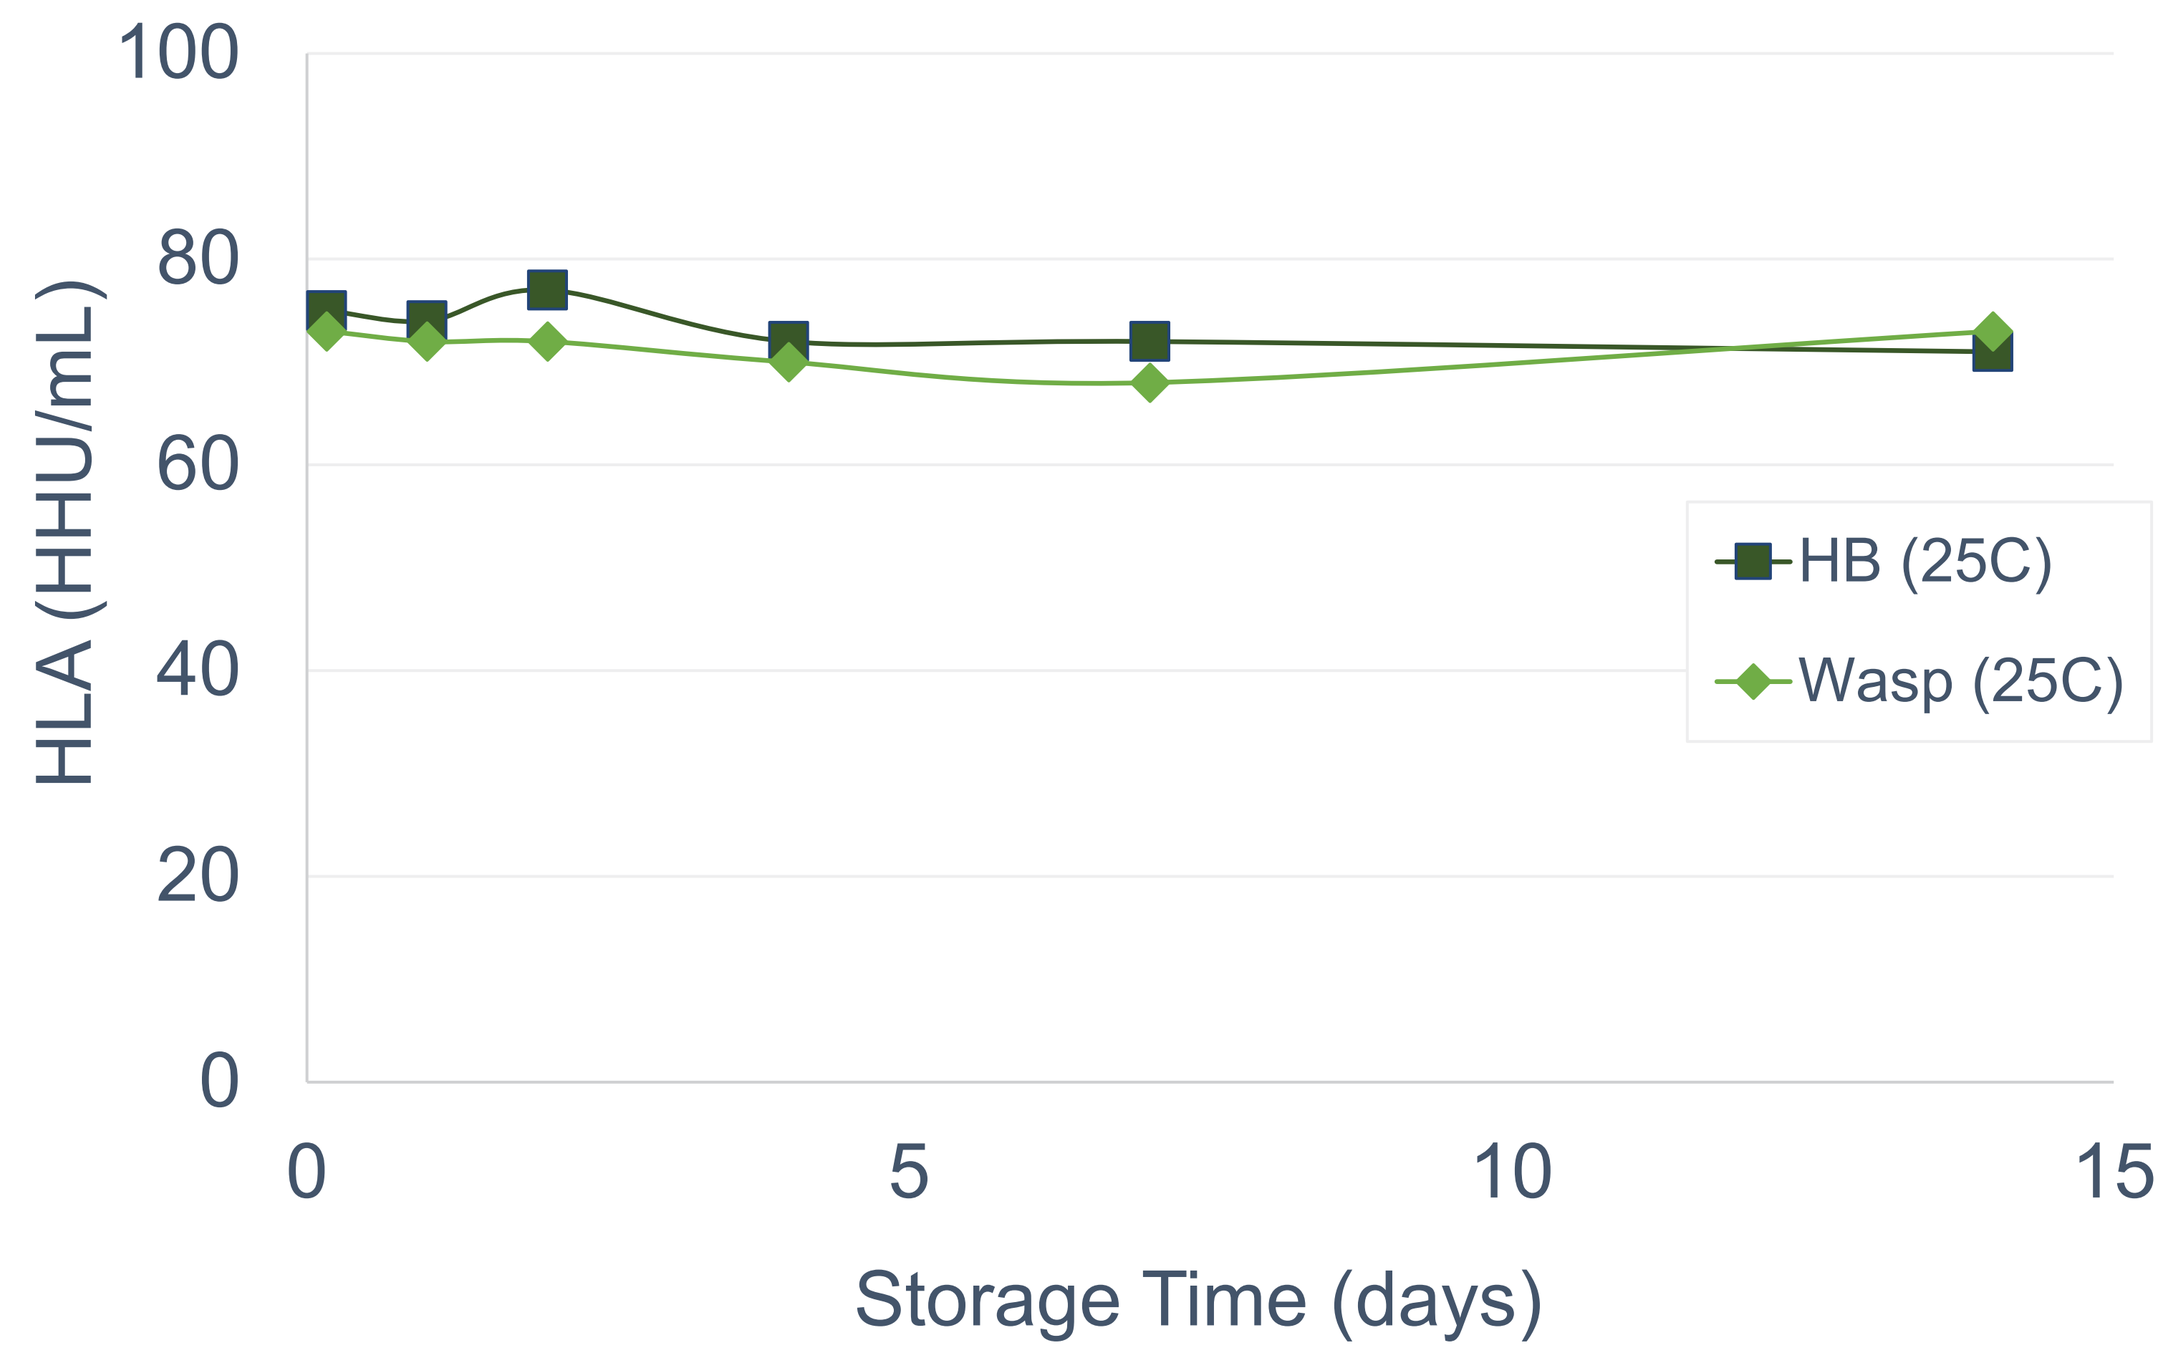
HLA (HHU/mL)
Storage Time (days)
HB (25C)
Wasp (25C)

Fig. 3 – PLA Activity for WFH and YJ

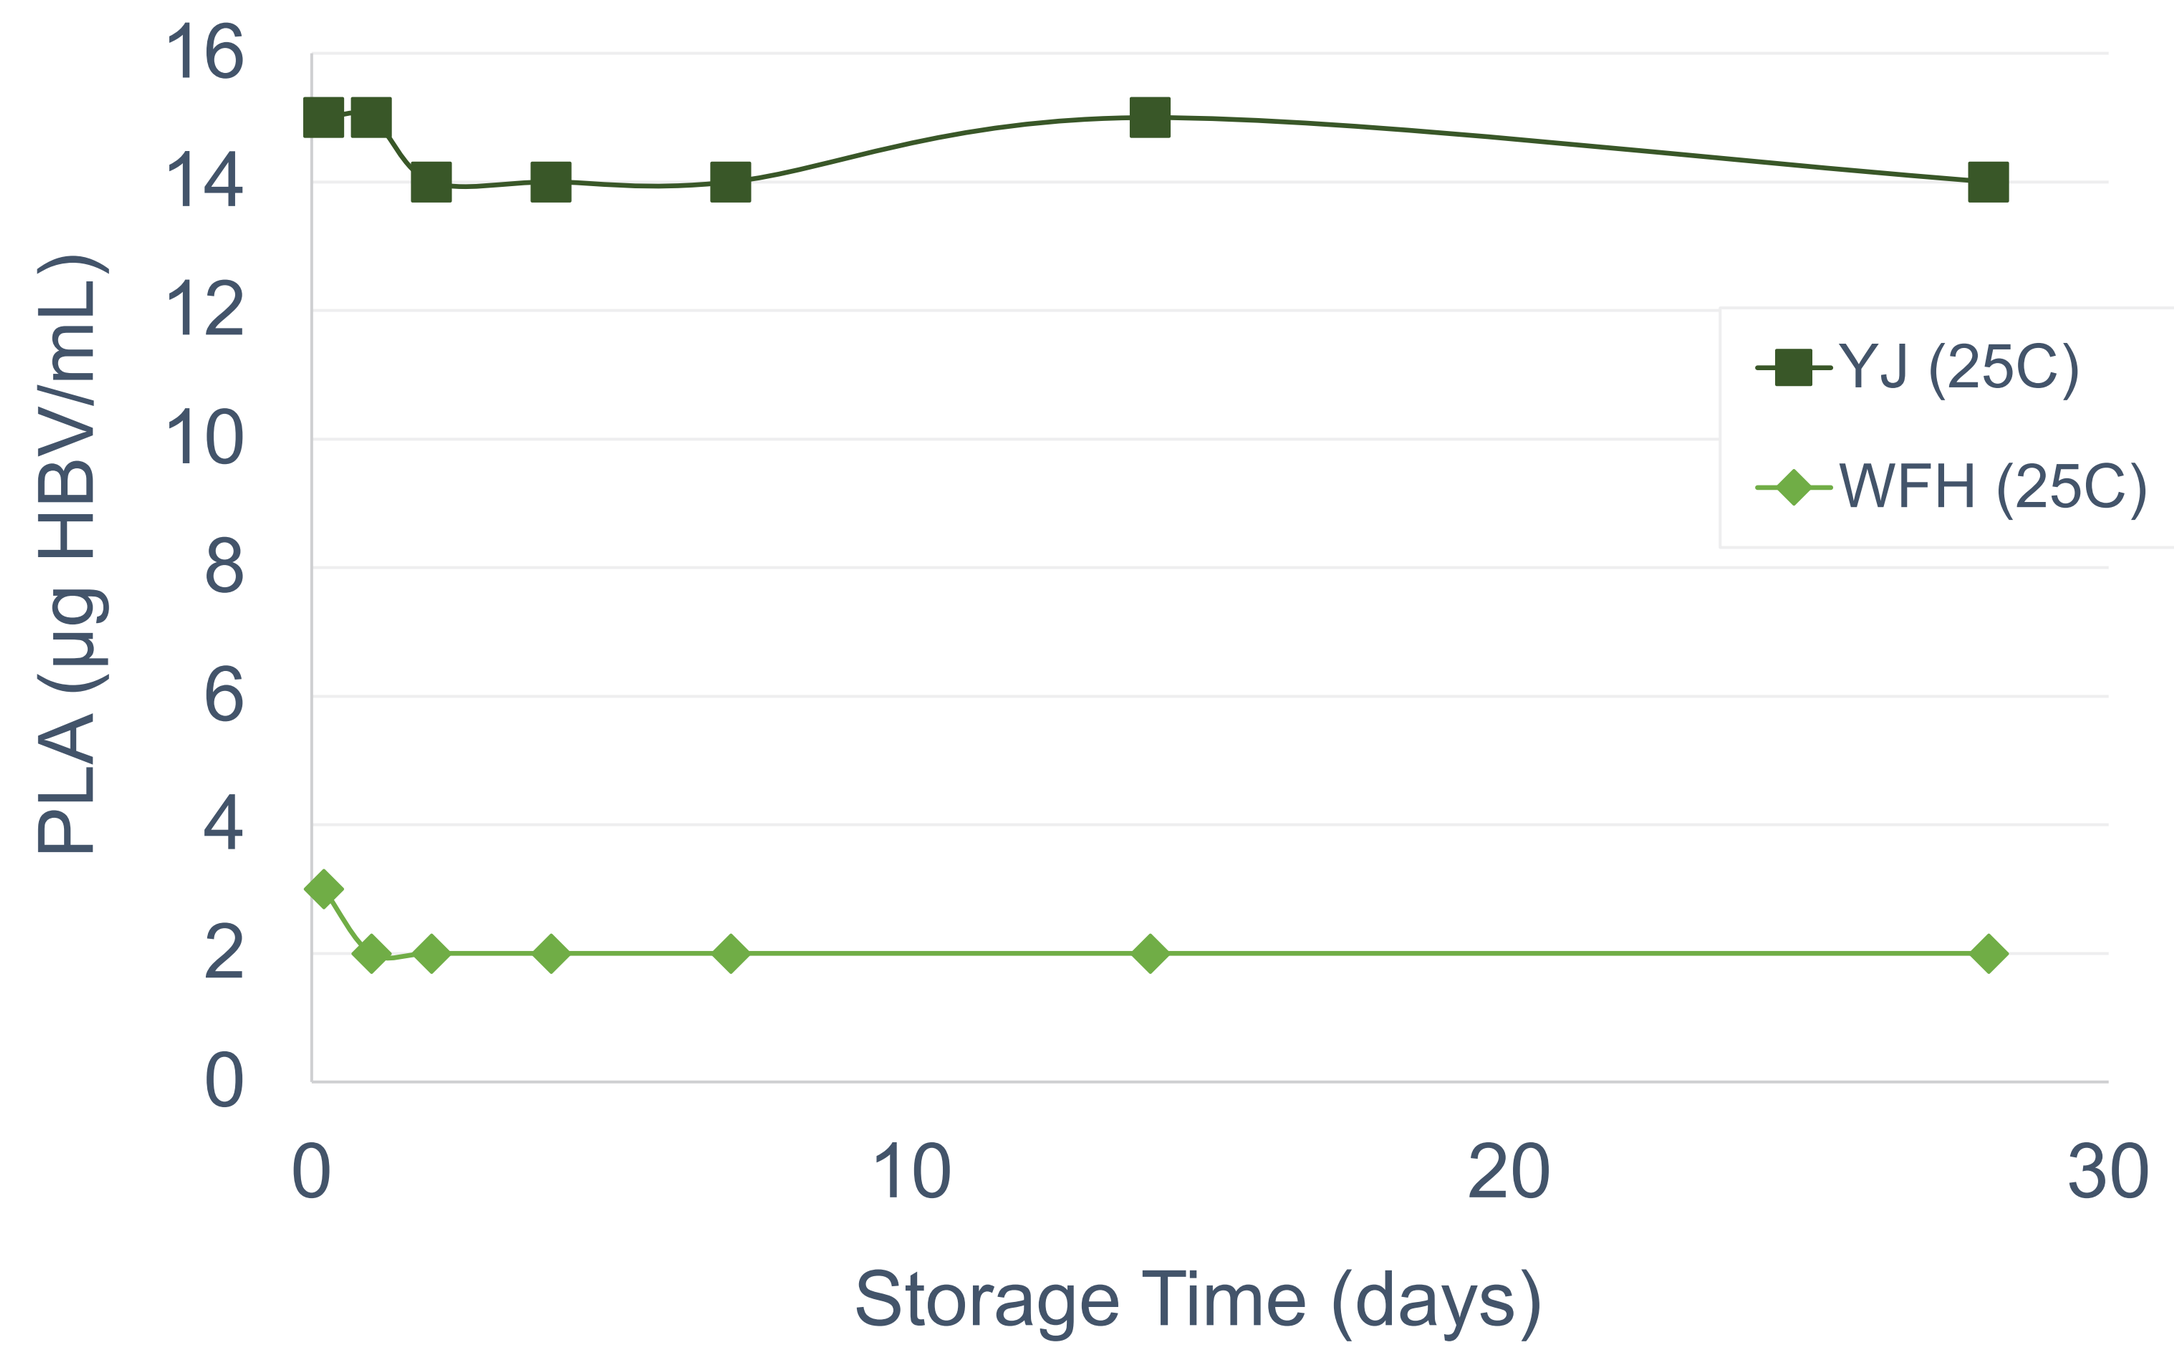
PLA (µg HBV/mL)
Storage Time (days)
YJ (25C)
WFH (25C)

Fig. 4 – HLA Activity for WFH and YJ

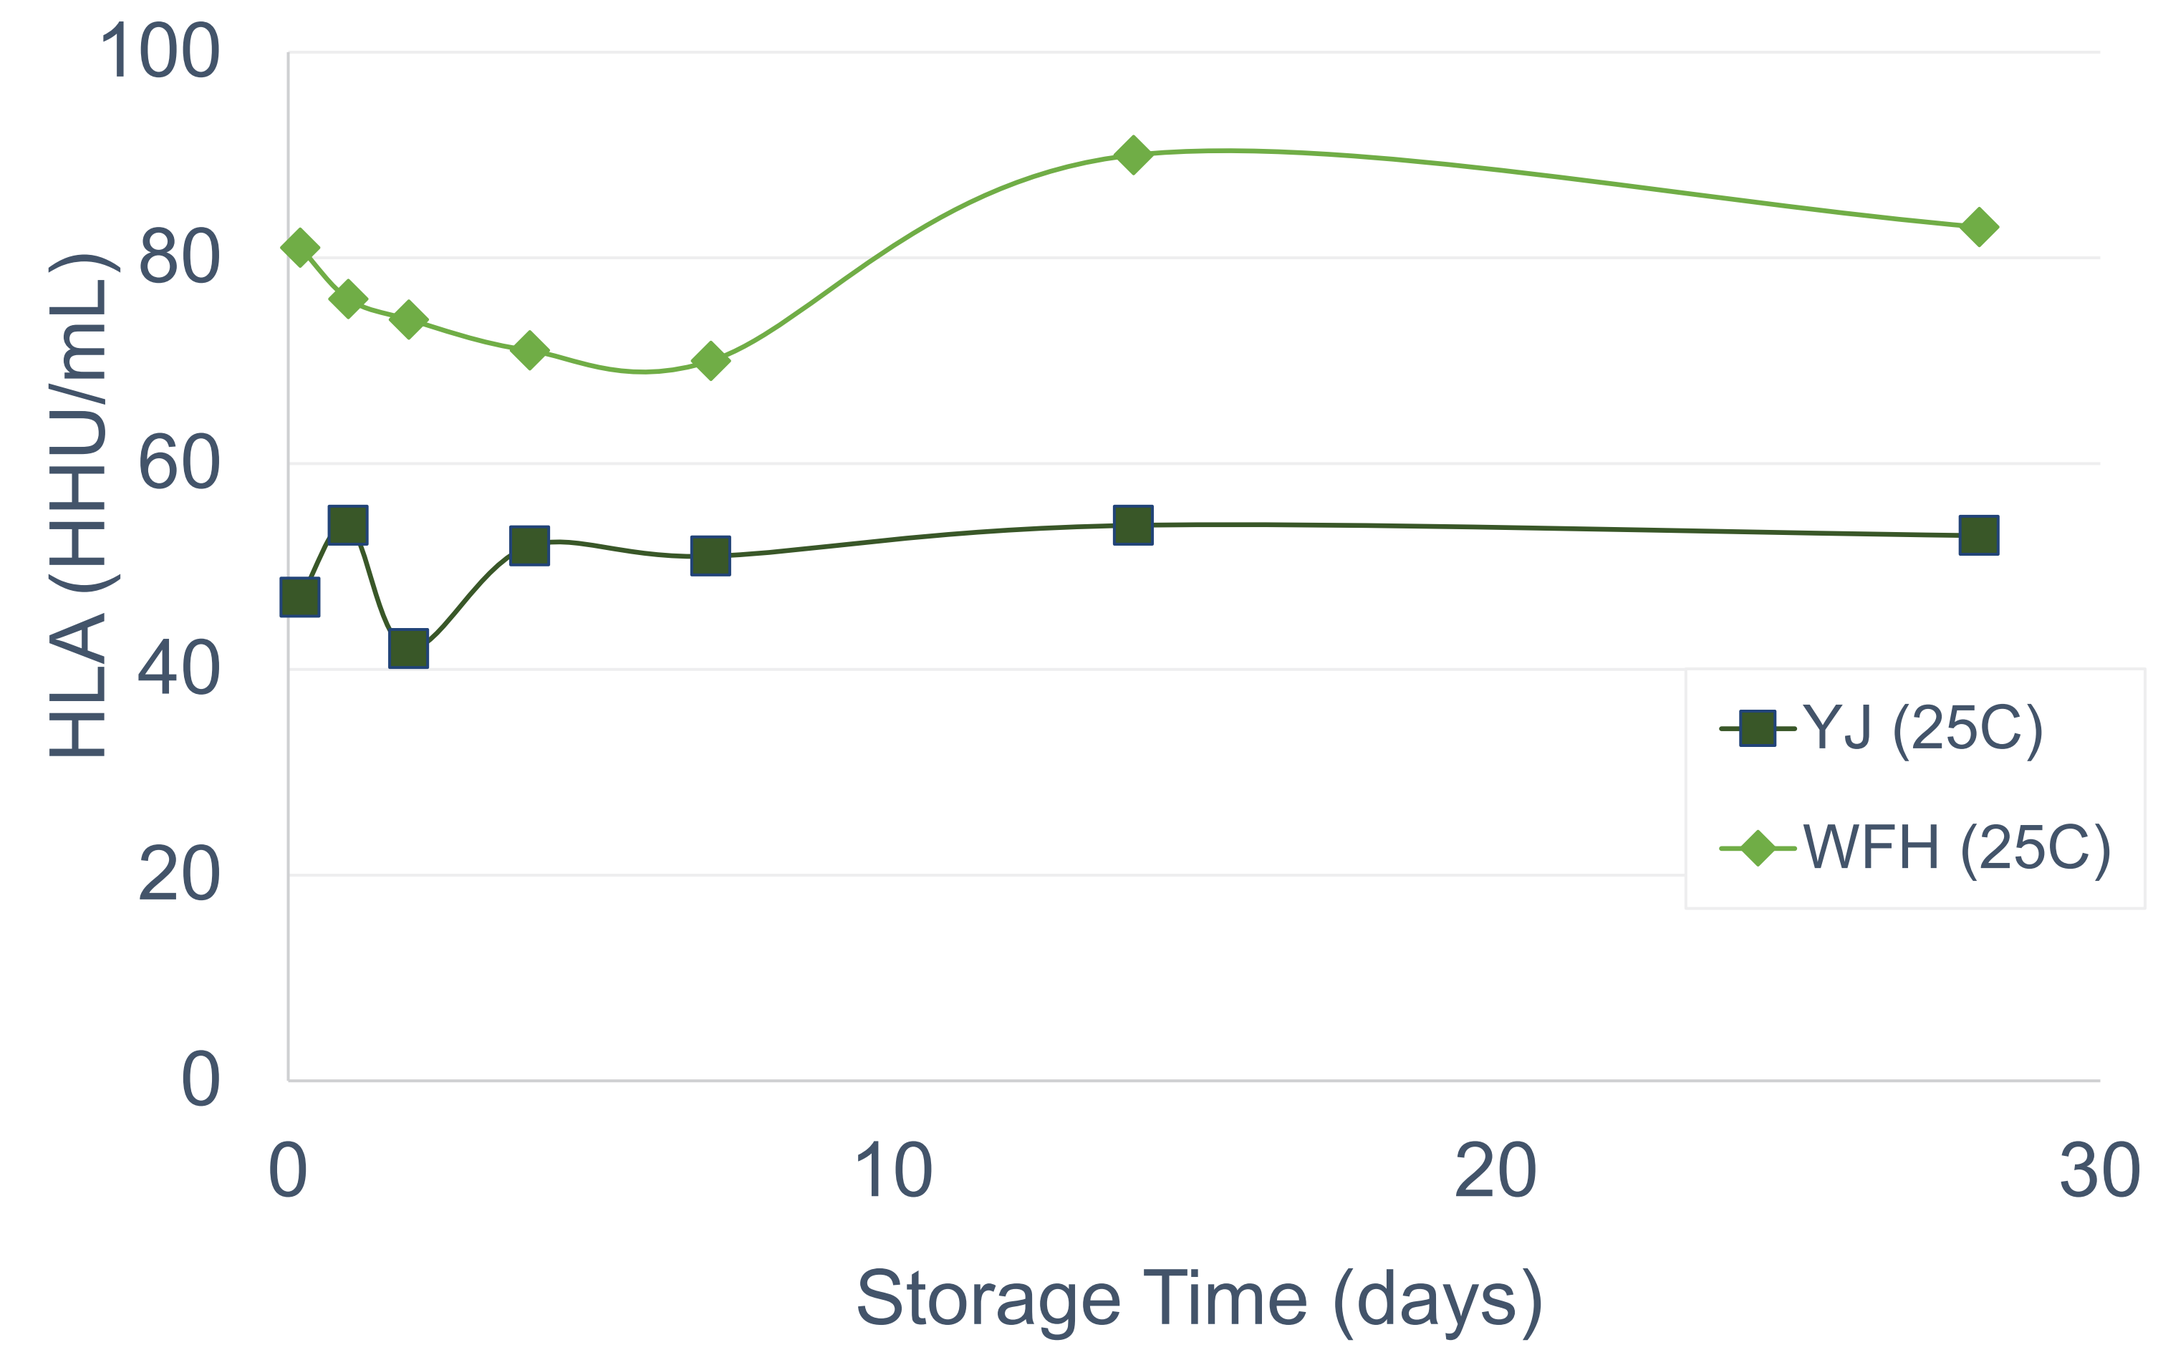
HLA (HHU/mL)
Storage Time (days)
YJ (25C)
WFH (25C)

Fig. 5 – PLA Activity for YH

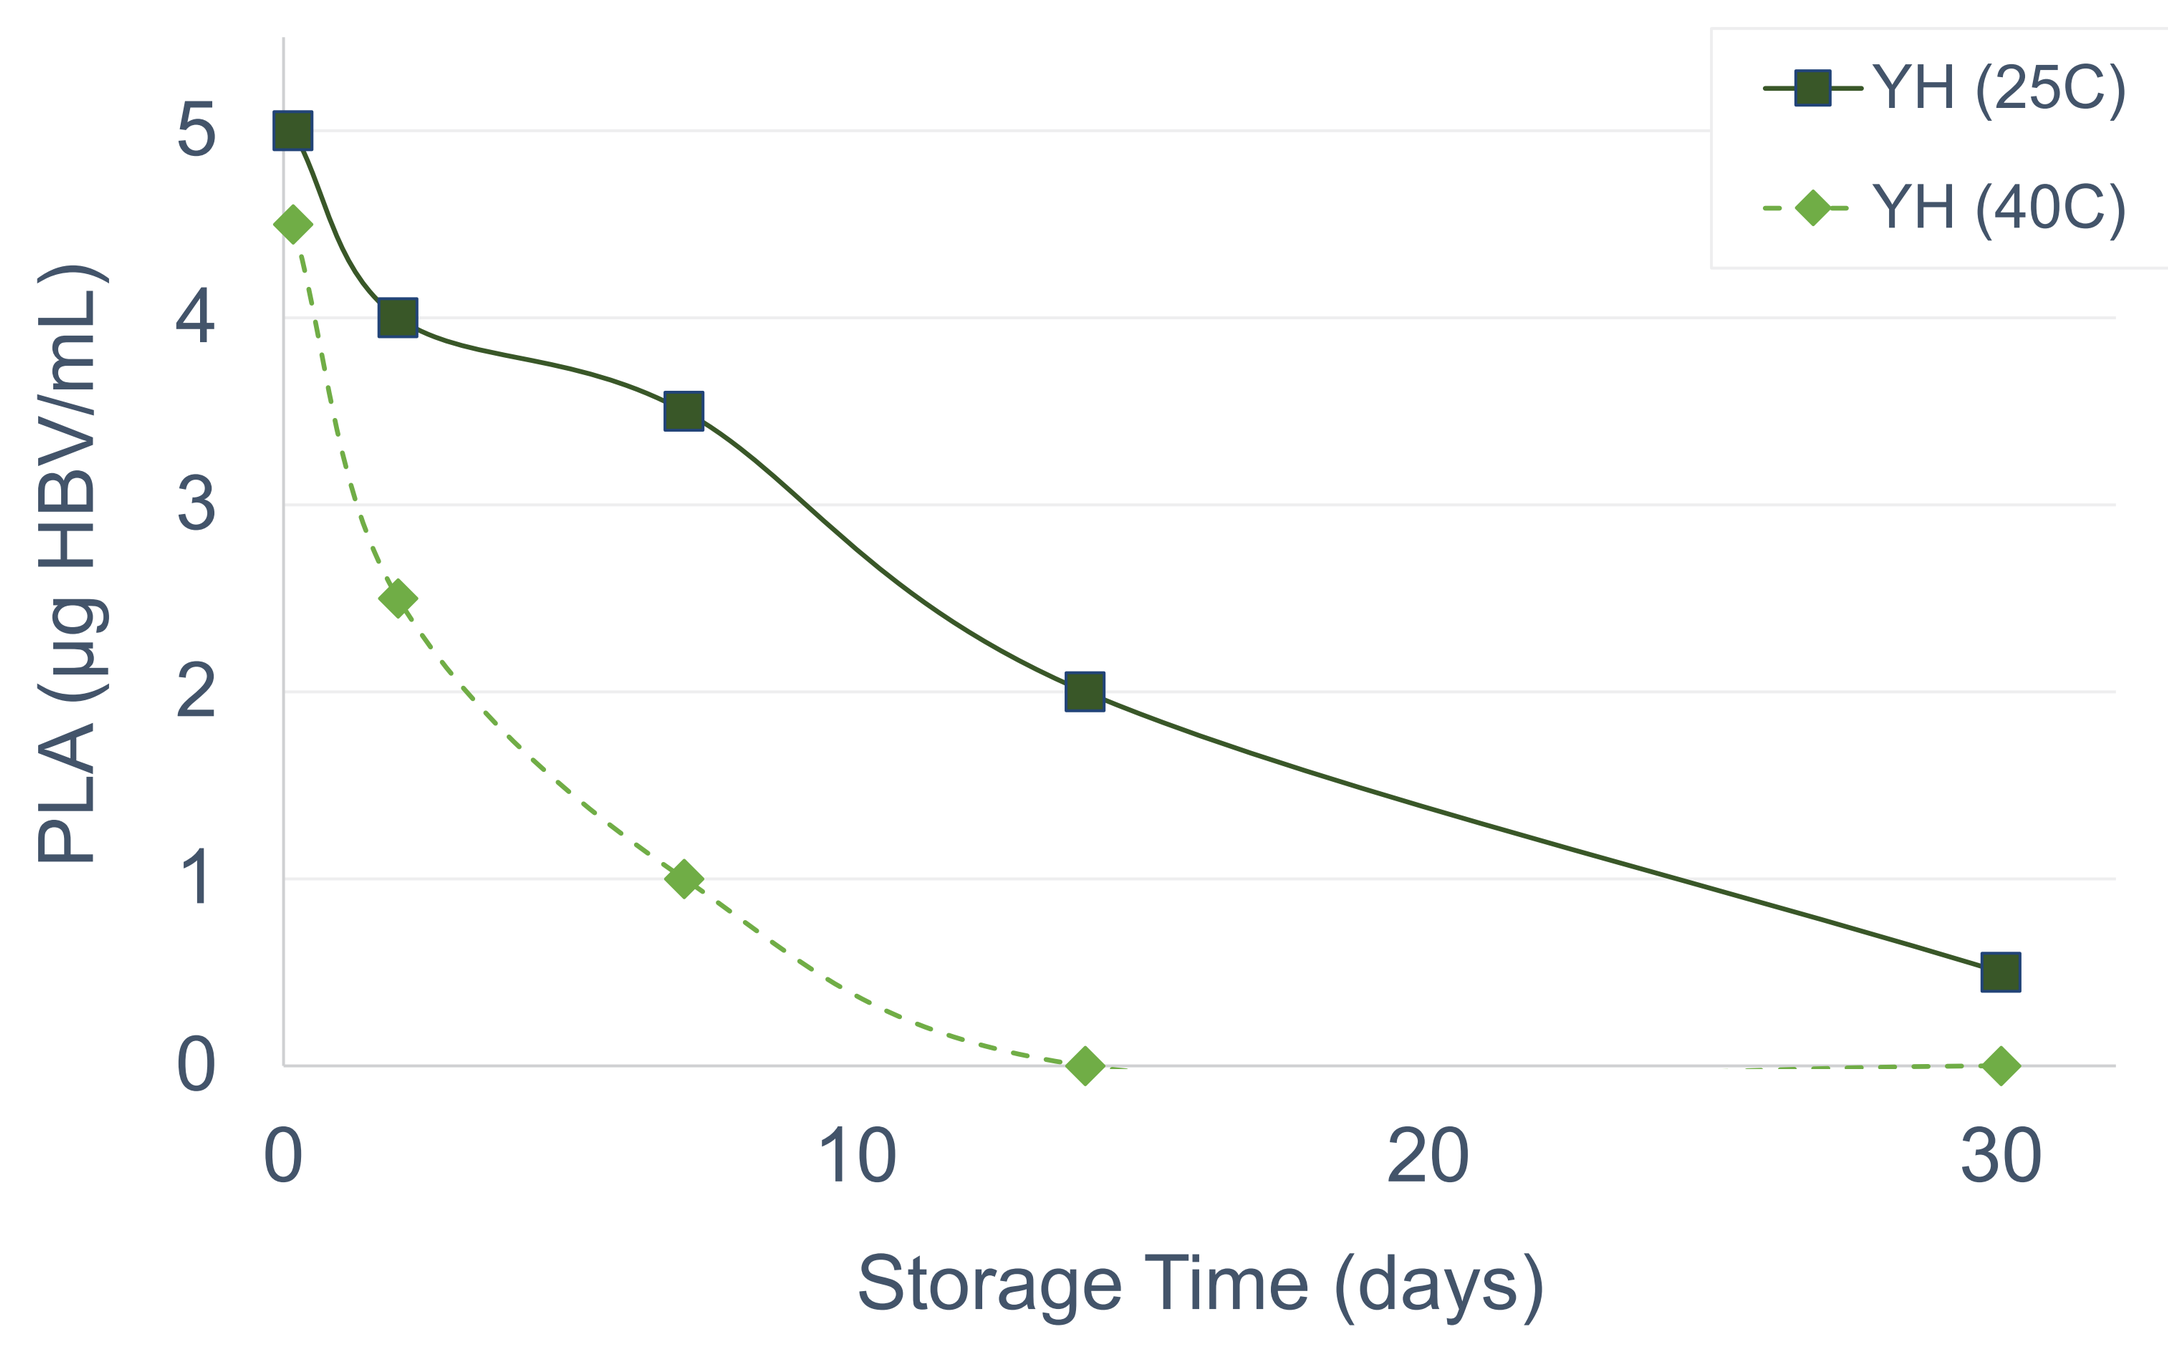
PLA (µg HBV/mL)
Storage Time (days)
YH (25C)
YH (40C)

Fig. 6 – HLA Activity for YH

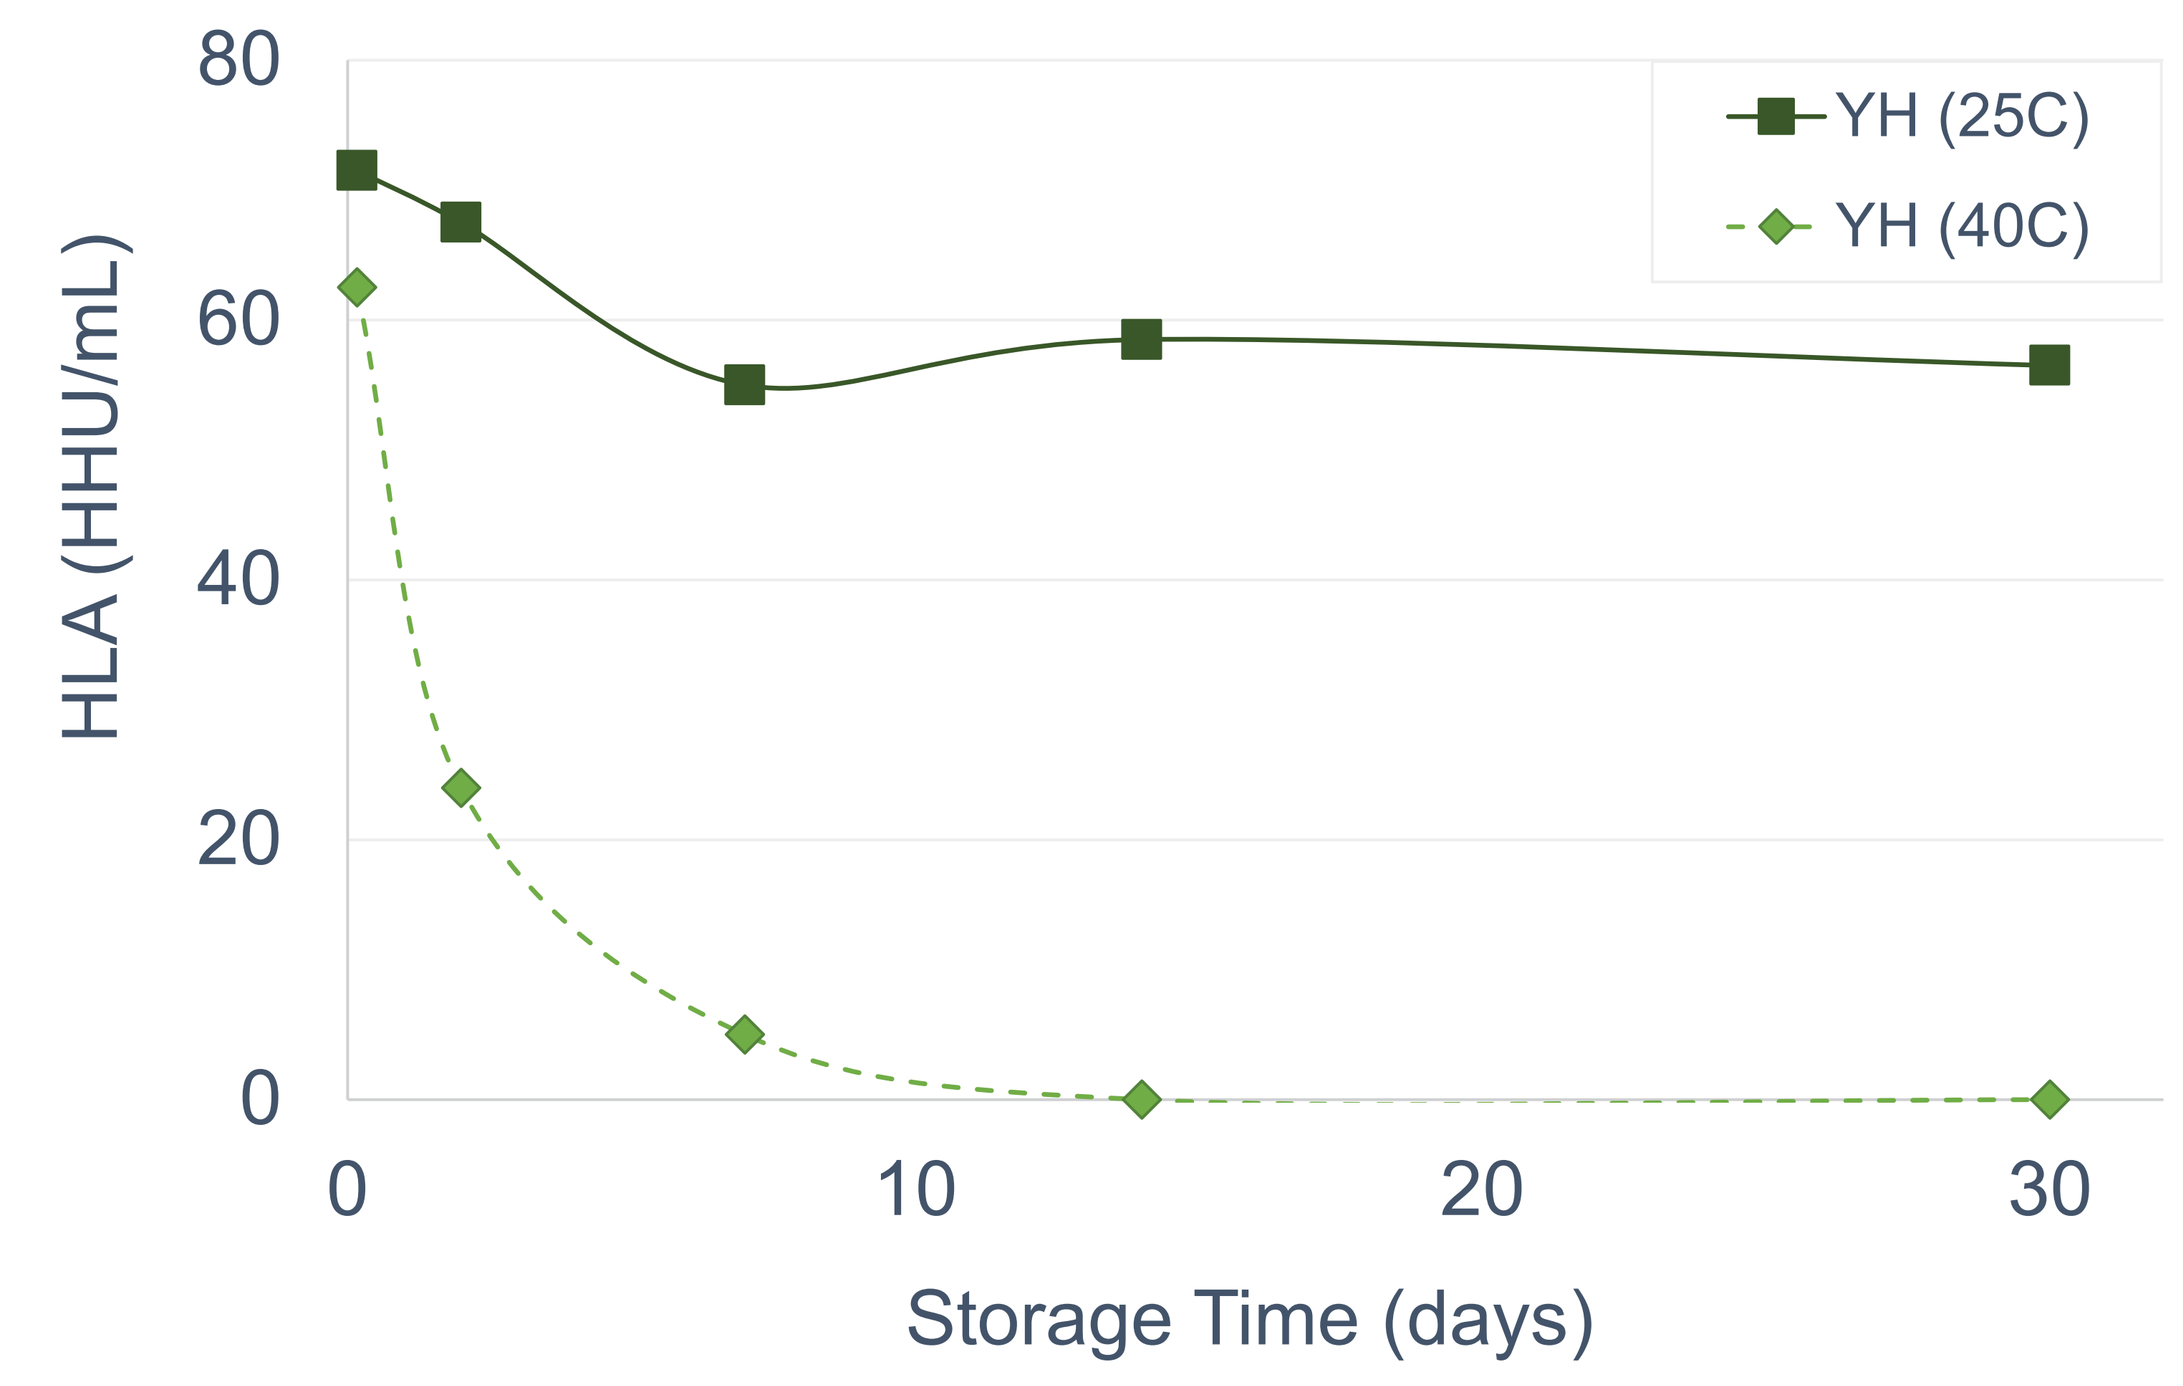
HLA (HHU/mL)
Storage Time (days)
YH (25C)
YH (40C)

PLA Reference Unit: µg Honey Bee Venom (HBV)/mL

HLA Reference Unit: Honey Bee Hyaluronidase Unit (HHU)/mL)

Discussion

Storage at Room Temperature (25°C):

- For Honey Bee and Wasp: PLA initially decreased ~23%, followed by continued gradual decline, but stayed above specification at 14 days. HLA was stable.
- For WFH and YJ: PLA and HLA remained stable for 28 days.
- For YH: PLA steadily decreased, with low or no activity detected at 30 days. HLA decreased ~23% at 7 days, then remained stable.

Storage at Elevated Temperature (40°C):

- PLA and HLA significantly decreased in YH, with no activity detected after 7-14 days.

For all reconstituted venoms, no bioburden was detected at the end of study.

Conclusions

Honey Bee, Wasp, White Faced Hornet, and Yellow Jacket products reconstituted to 100 µg/mL remain within stability specifications after exposure to room temperature conditions for 14-28 days. However, exercise caution when considering use of Yellow Hornet that has been at room temperature for more than 7 days or exposed to elevated temperatures for more than a day.